The Anti-Tumor Activity of IFNβ and Membrane-Stable CD40L Expressing Oncolytic Virus MEM-288 in NSCLC Patients is Associated with Modulation of the Tumor Microenvironment and Systemic Immune Response

Andreas N Saltos[1], Christy Arrowood[2], Georgia Beasley[2], James Ronald[2], Ghassan El-Haddad[1], Jhanelle E. Gray[1], Bruna Pellini[1], Tawee Tanvetyanon[1], Luiziane Guerra-Guevara[1], Uzma Khan[2], Steven Wolf[2], Lin Gu[2], Xiaofei F. Wang[2], Hong Zheng[1], Dana Foresman[1], Xiaoqing Yu[1], Mark J Cantwell[3], Scott J Antonia[2], Amer A Beg[1], Neal E Ready[2]

[1]Moffitt Cancer Center, Tampa, Florida, USA; [2]Duke Cancer Institute, Durham, North Carolina, USA; [3]Memgen, Inc, Houston, Texas, USA

Background

- MEM-288 is a conditionally-replicative oncolytic adenovirus (Ad5) encoding transgenes for human interferon beta (IFNβ) and a recombinant membrane-stable form of CD40-ligand (MEM40), two potent activators of the immune response and cDCs.

MEM-288 design and backbone

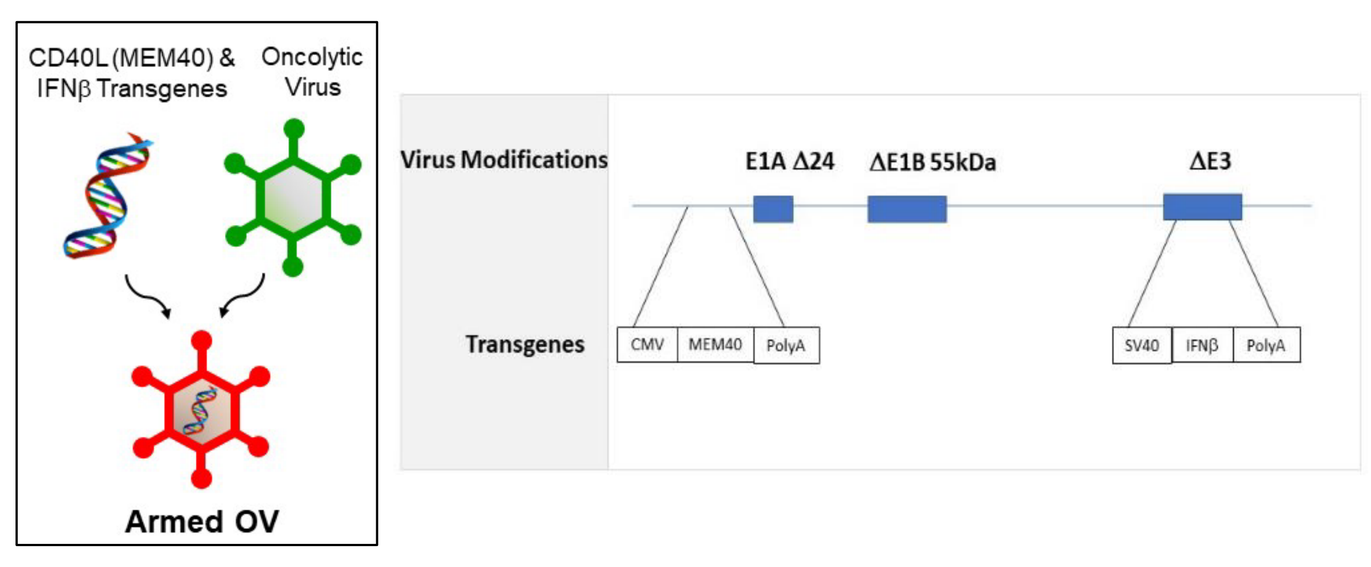

CD40L (MEM40) & IFNβ Transgenes
Oncolytic Virus
Armed OV
Virus Modifications
E1A Δ24
ΔE1B 55kDa
ΔE3
Transgenes
CMV
MEM40
PolyA
SV40
IFNβ
PolyA

- In preclinical studies, MEM40 and IFNβ in situ co-expression induced higher conventional dendritic cell (cDC) activation than either molecule alone, led to a dramatic increase in lymph node migration, a systemic anti-tumor CD8+ T cell response, and regression of established tumors in a manner dependent upon type 1 cDCs (Zheng et al., Cancer Immunol Res. 2023).
- MEM40 and IFNβ expression enhanced generation of both Granzyme B+ CD8+ T cell effectors as well as TCF1+ stem-like CD8+ T cells that are known to be strongly associated with response to immune checkpoint inhibitors (ICIs). Intralesional therapy with MEM40 and IFNβ expressing adenovirus synergized with ICIs, leading to effective control of distant tumors and lung metastases.
- MEM-288 is being evaluated in an open-label, multi-center, phase 1 clinical trial (NCT05076760). Here, we present results of the completed first part of the trial, a monotherapy dose-escalation of MEM-288 in patients with select solid tumors.

Clinical Trial Design

- Phase 1 dose-escalation, multi-center, open-label clinical trial (NCT05076760) using a BOIN design to investigate intratumoral administration of MEM-288 in patients with select solid tumors
- The primary objective was to determine the MTD and recommended phase II dose of MEM-288 monotherapy across 3 dose levels (DL) spanning 1e10 to 1e11 viral particles by intratumoral injection once every 3 weeks, for up to 6 injections. Secondary objectives include assessment of efficacy including response rate of injected and non-injected tumors assessed separately.
- Secondary exploratory analyses evaluated anti-tumor immune responses.

Key Eligibility criteria:
- Advanced NSCLC, cutaneous squamous or Merkel cell carcinoma, melanoma, pancreatic cancer, triple-negative breast cancer, or head and neck cancer which is refractory to standard therapies including anti-PD(L)1.
- Tumor lesion deemed feasible for biopsy and MEM-288 intratumoral injection

- Patient tumor biopsies were collected prior to the first and 2nd MEM-288 injections. We collected peripheral blood at serial timepoints.

Study design

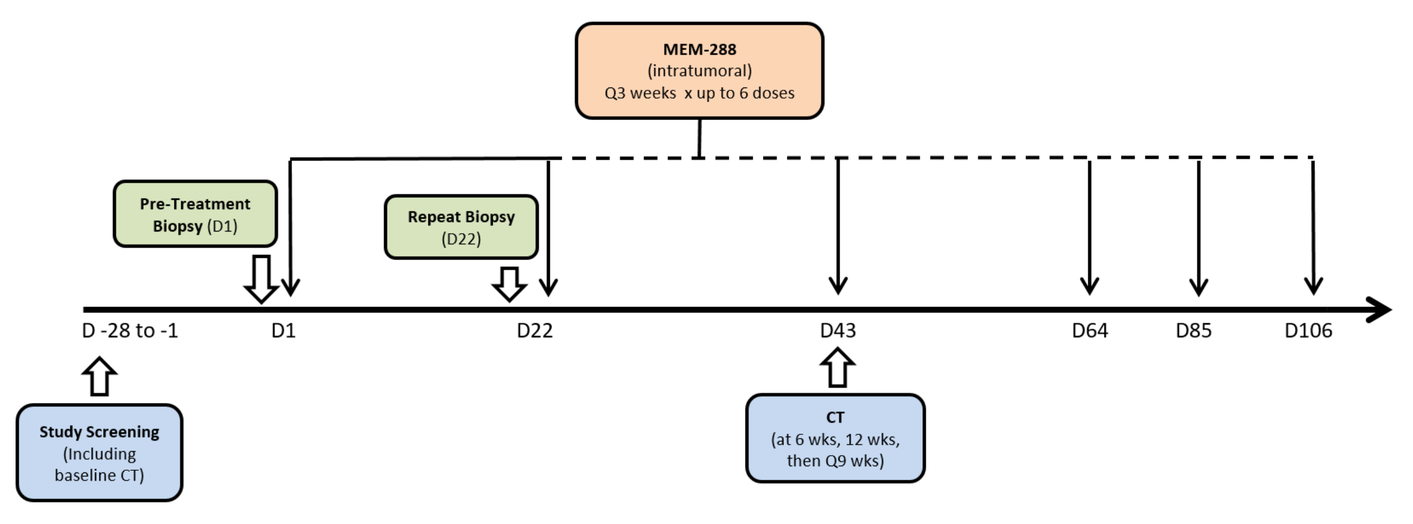

MEM-288 (intratumoral) Q3 weeks x up to 6 doses
Pre-Treatment Biopsy (D1)
Repeat Biopsy (D22)
D -28 to -1
D1
D22
D43
D64
D85
D106
Study Screening (Including baseline CT)
CT (at 6 wks, 12 wks, then Q9 wks)

Patient Demographics

Age (years)	Previous Systemic Therapies
Median: 64 Range: 39-76	Median: 4 Range: 1-7
Sex	**Brain Metastases (Treated) at Baseline**
Male 60% Female 40%	Yes 33% No 67%
Smoking History	**Histologic Cancer Type**
Current or Former: 60% Never 27% Undisclosed 13%	NSCL Adenocarcinoma 80% NSCLC Squamous Cell 7% NSCLC Large Cell Neuroendocrine 7% Pancreatic 7%

- 14 patients with metastatic NSCLC refractory to anti-PD(L)1 and platinum chemotherapy received MEM-288 treatment

Safety

Treatment Related AEs

Adverse Reactions	Grade 1 n (%)	Grade 2 n (%)	Total n (%)
Injection Site Pain/Tenderness	2 (13%)	1 (7%)	**3 (20%)**
Abdominal pain	1 (7%)	1 (7%)	**2 (13%)**
Chills	2 (13%)	0	**2 (13%)**
Fever	2 (13%)	0	**2 (13%)**
Constipation	0	1 (7%)	**1 (7%)**
Headache	0	1 (7%)	**1 (7%)**
Nausea	1 (7%)	0	**1 (7%)**
Fatigue	1 (7%)	0	**1 (7%)**
Injection site itch	1 (7%)	0	**1 (7%)**
Dehydration	1 (7%)	0	**1 (7%)**
Myalgia	1 (7%)	0	**1 (7%)**

Safety Conclusions

- Side effects consisted primarily of injection site reactions and flu-like symptoms, expected for viral immunotherapy.
- No DLTs occurred, and no patients discontinued treatment due to toxicity.
- There were no Grade ≥3 TRAEs.
- 1x10^{11} viral particles was selected as the recommended phase 2 dose for clinical expansion studies.

Efficacy and Anti-Tumor Activity

Patient ID# (n=14)[1]	NSCLC Histologic Cancer Type	MEM-288 Dose Level	Injected Lesion Description	Non-Injected Lesions Descriptions	ORR	Injected Lesion (RR)	Non-Injected Lesions (RR)
002-001	Adenocarcinoma	Low	Right wall chest mass	Left subpectoral lymphadenopathy, precranial lymphadenopathy	PD	PR	PD
002-002	Adenocarcinoma	Low	Left cervical lymphadenopathy	Right hilar lymphadenopathy	SD	PR	SD
001-001	Adenocarcinoma	Low	Gluteus maximus (Pelvis)	Supraclavicular lymph node, pleura, mediastinal lymph node, muscle	PD	PD	PD
001-002	Adenocarcinoma	Mid	Liver	Pleura, liver, celiac lymph node	SD	PD	SD
002-003	Adenocarcinoma	Mid	Left lateral neck adenopathy	Left neck adenopathy, thoracic adenopathy	PD	PD	PD
002-005	Large Cell Neuroendocrine	Mid	Liver	Subcarinal lymph node, portacaval lymph node	PD	PR	PD
001-005	Adenocarcinoma	Mid	Lung left upper lobe	Lung upper right lobe	PD	PD	PD
002-006[2]	Adenocarcinoma	High	Right liver lobe	Anteromedial pleural, lower pulmonary lobe, subcarinal node, right liver lobe	PD	-	-
001-004	Adenocarcinoma	High	Lung left upper lobe	Pleura, mediastinal lymph node	SD	PD	SD
002-007	Adenocarcinoma	High	Anterior left axillary lymph node	Left perihilar, anterior left axillary lymph node	PD	SD	PD
002-008	Squamous Cell	High	Extracranial left temporal mass	Left apical lung, left lower lobe, subcarinal lymph node	PD	PD	SD
002-009	Adenocarcinoma	High	Hepatic segment 5 mass	Right lower lobe pulmonary mass, hepatic segment 2/3 mass	PD	PD	PD
002-010[2]	Adenocarcinoma	High	Left axillary lymphadenopathy	Subcarinal lymphadenopathy, left subpleural/intercostal implant, right upper lob pulmonary mass	PD	-	-
002-012	Adenocarcinoma	High	Left supraclavicular lymph node	Left abdominal para-aortic lymphadenopathy	PR	PR	Non CR/PD
				DCR %	29% (n=4)	36% (n=5)	36% (n=5)
				Clinical Activity %[3]	50% (n=7)		

[1]ID# prefix 001=Duke and 002=Moffitt
[2]Discontinued study prior to scheduled 6-week CT scans due to clinical progression
[3]Clinical Activity defined as patients with RECIST 1.1 objective response of SD/PR **and/or** measurable shrinkage of injected lesion

Tumor Shrinkage (Injected Tumor)

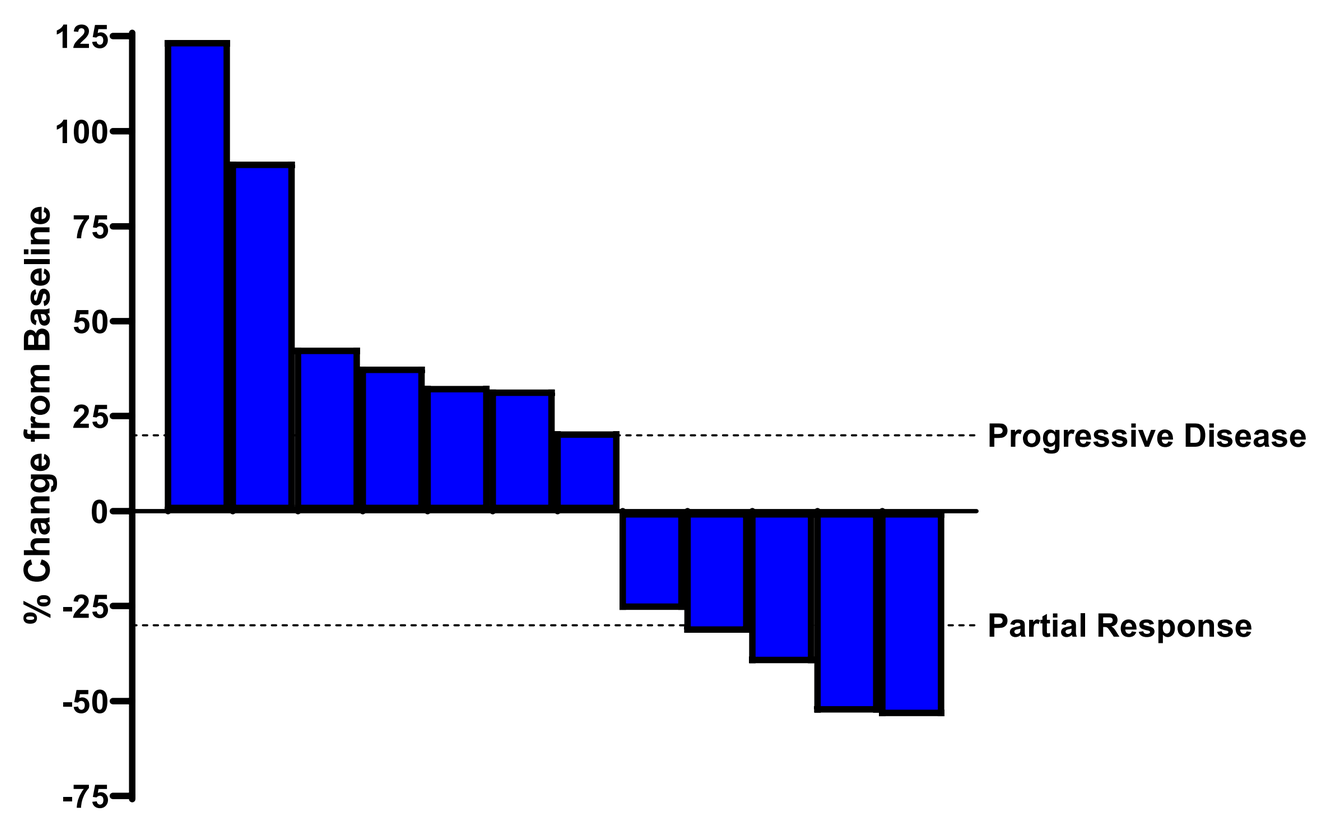

% Change from Baseline
125
100
75
50
25
0
-25
-50
-75
Progressive Disease
Partial Response

Injected Tumor Response Correlates to # of Prior Lines of Therapy

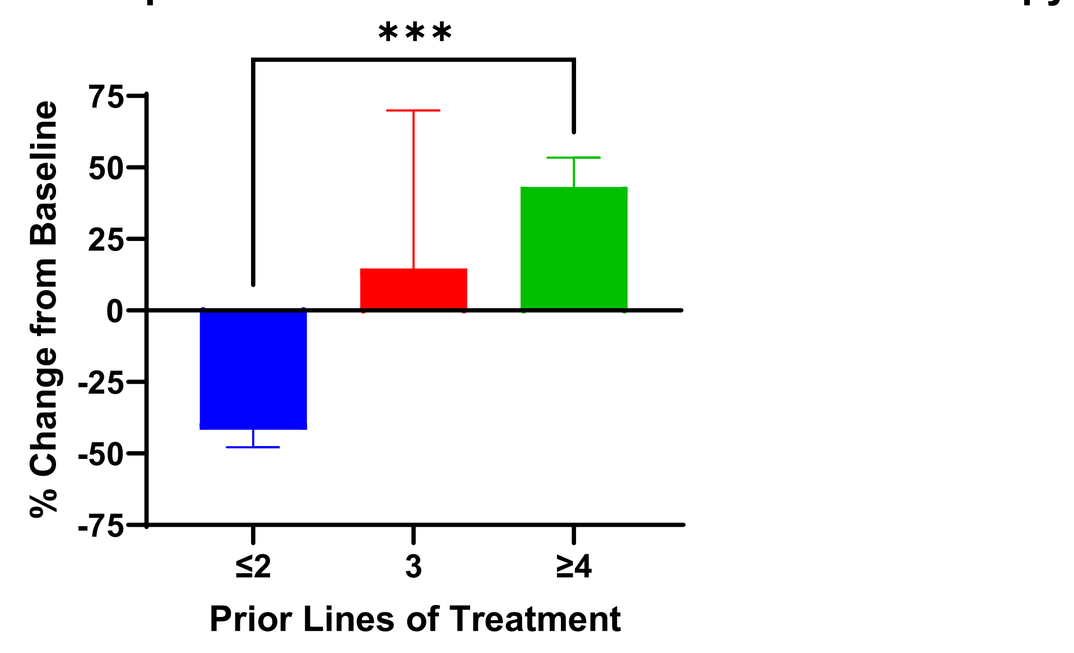

% Change from Baseline
75
50
25
0
-25
-50
-75
≤2
3
≥4
Prior Lines of Treatment

Test for linear trend indicates a significant trend (P=0.0394) of injected tumor response to prior treatment lines

Efficacy Conclusions

- MEM-288 was successfully injected into superficial (n=7) and deep visceral tumors (n=7)
- Anti-tumor activity was primarily seen in injected tumors, and to a lesser extent in abscopal lesions. 42% of evaluable NSCLC patient (5 of 12) showed tumor shrinkage following MEM-288 treatment.
- 50% of NSCLC patients showed MEM-288-mediated clinical activity: patients with RECIST 1.1 objective response of SD/PR **and/or** measurable shrinkage of injected lesion
- MEM-288 injected tumor shrinkage was limited to the 5 patients with ≤3 prior lines of treatment
- Clinical activity was seen across all dose levels of MEM-288

Notable Patient Response Outcomes

Patient 002-001 - MEM-288 Tumor Shrinkage Followed by Chemotherapy Rechallenge Durable Response

Time from MEM-288 Trx Initiation (months)	0 mos	1.5 mos	3 mos	5 mos	17 mos
Prior Line of Therapy 1st: Osimertinib 2nd: Carboplatin paclitaxel bevacizumab atezolizumab	MEM-288 Pre-Trx	2 Cycles MEM-288 Completed (53% shrinkage of injected lesion)	2 Cycles docetaxel plus ramucirumab (taxane/anti-VEGF rechallenge response)	7 Cycles docetaxel plus ramucirumab (Complete response and systemic therapy break months 5-11)	Radiotherapy and osimertinib rechallenge at 11 mos for brain mets (Ongoing extracranial complete response)
Injected Lesion Subcutaneous chest wall	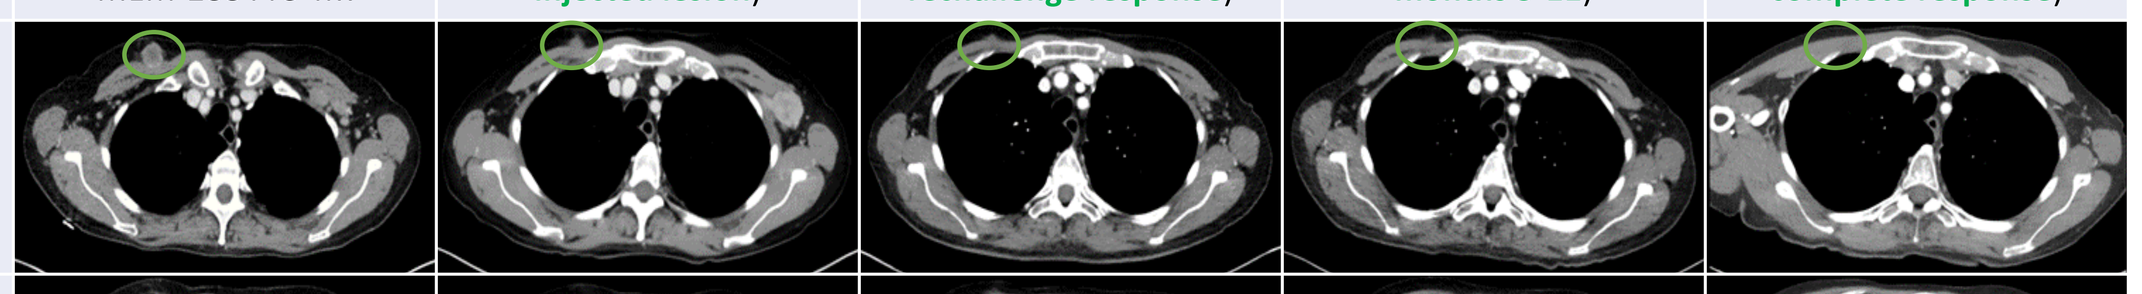				
Non-Injected Target Lesion Left axillary lymph node	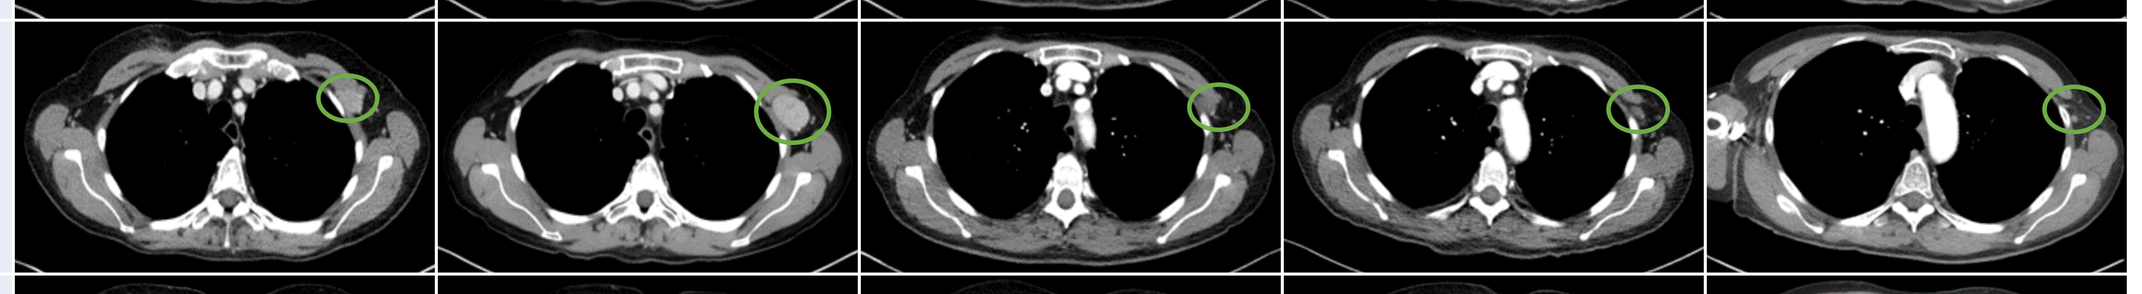				
Non-Injected Target Lesion Mediastinal, hilar lymph node	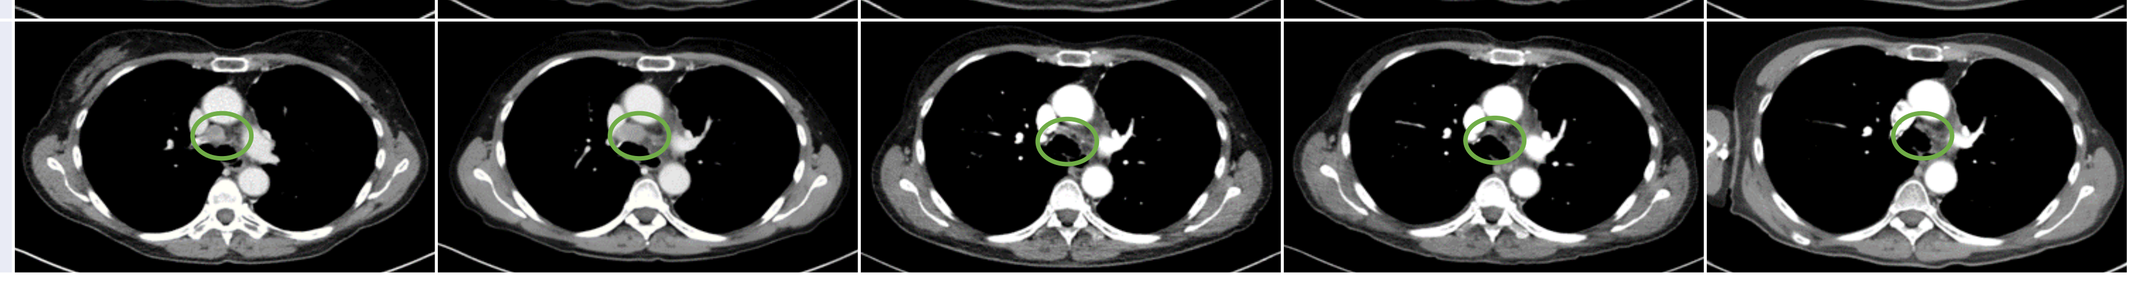				

Patient 002-005 - MEM-288 Tumor Shrinkage Followed by Chemotherapy Rechallenge Durable Response

Time from MEM-288 Trx Initiation (months)	0 mos	1.5 mos	7 mos
Prior lines of trx 1st: Carboplatin etoposide 2nd: Atezolizumab	MEM-288 Pre-Trx	2 Cycles MEM-288 Completed (40% shrinkage of injected lesion with calcification of lesions)	6 Cycles carboplatin plus etoposide rechallenge (PR achieved after 2 cycles)
Injected Lesion Inferior liver metastasis	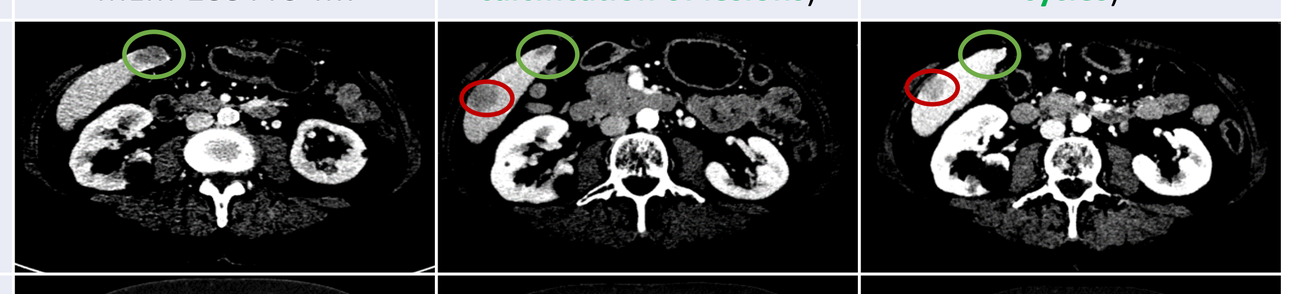		
Non-Injected Target Lesion Left periaortic lymph node	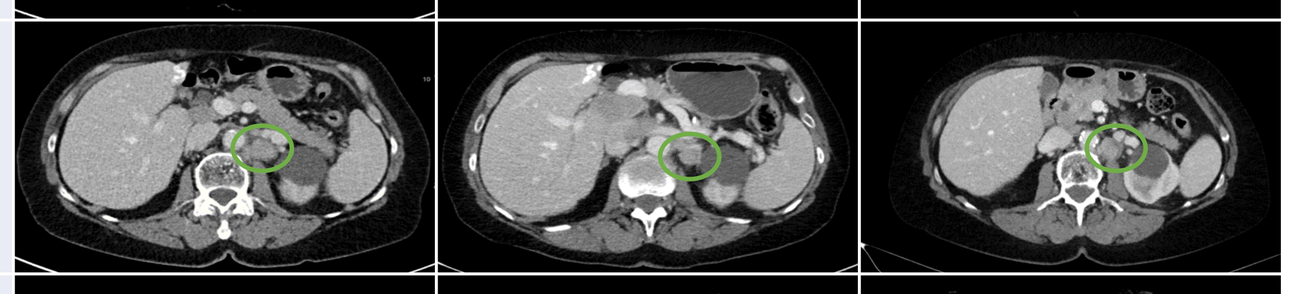		
Non-Injected Target Lesion Segment 5 liver metastases	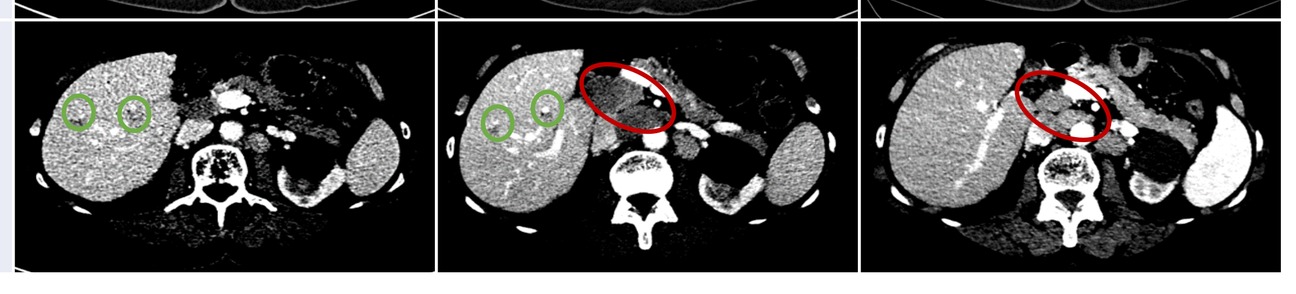		

Patient 002-012 – Pseudoprogression Followed by MEM-288 Tumor Shrinkage and Partial Response

Time from MEM-288 Trx Initiation (months)	0 mos	1.5 mos	3 mos	4 mos
Prior lines of trx 1st: Carboplatin pemetrexed pembrolizumab	MEM-288 Pre-Trx	2 Cycles MEM-288 (Pseudoprogression)	4 Cycles MEM-288 ORR = PR (32% shrinkage of injected lesion; non-CR/PD non-injected lesion)	6 Cycles MEM-288 completed
Injected Lesion Supraclavicular lymph node	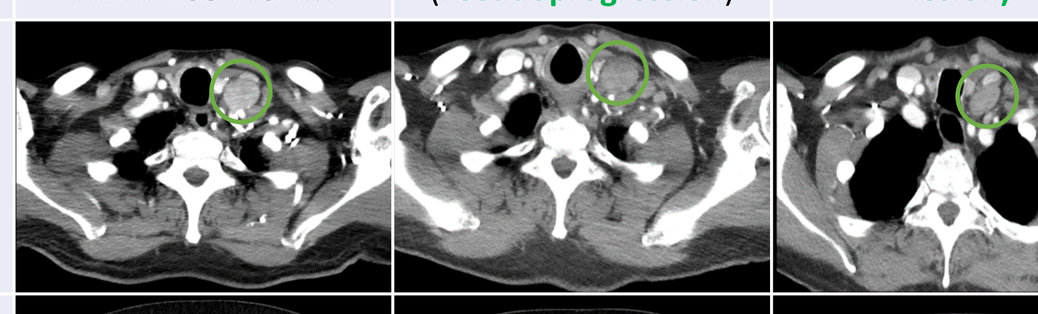			
Non-Injected Lesion Para-aortic lymph node	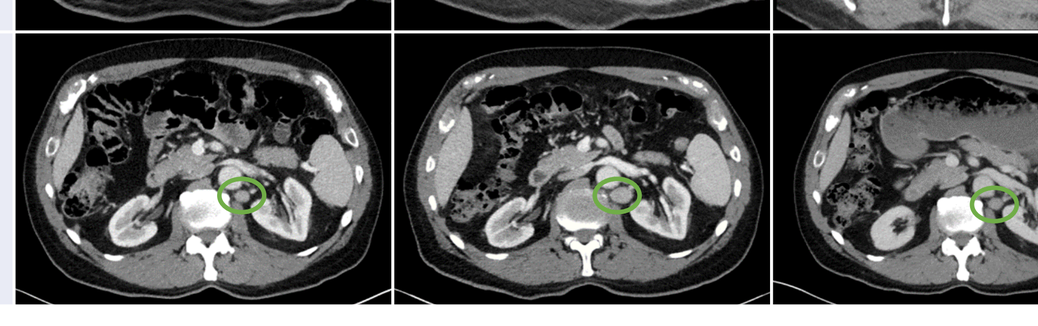			

Patient Response Conclusions

- Two patients developed subsequent durable responses, 1 CR and 1 PR, to salvage chemotherapy rechallenge.
- Patient 002-012 initially showed pseudoprogression following the first two MEM-288 injections. Continued MEM-288 injections resulted in tumor shrinkage and a RECIST 1.1 partial response.
- All responding patients showed signature biomarker and immune activation profiles
- Patient 002-012 mirrors the inclusion criteria for the next stage of this Phase 1 trial, an expansion study of MEM-288 with nivolumab for second-line treatment of advanced NSCLC refractory to front-line anti-PD(L)1 +/- chemotherapy

Biomarkers

Pre-Treatment TME Cell Subsets Identified as Potential Biomarkers of Response to MEM-288 Treatment

T Cells | cDC1 | Macrophages

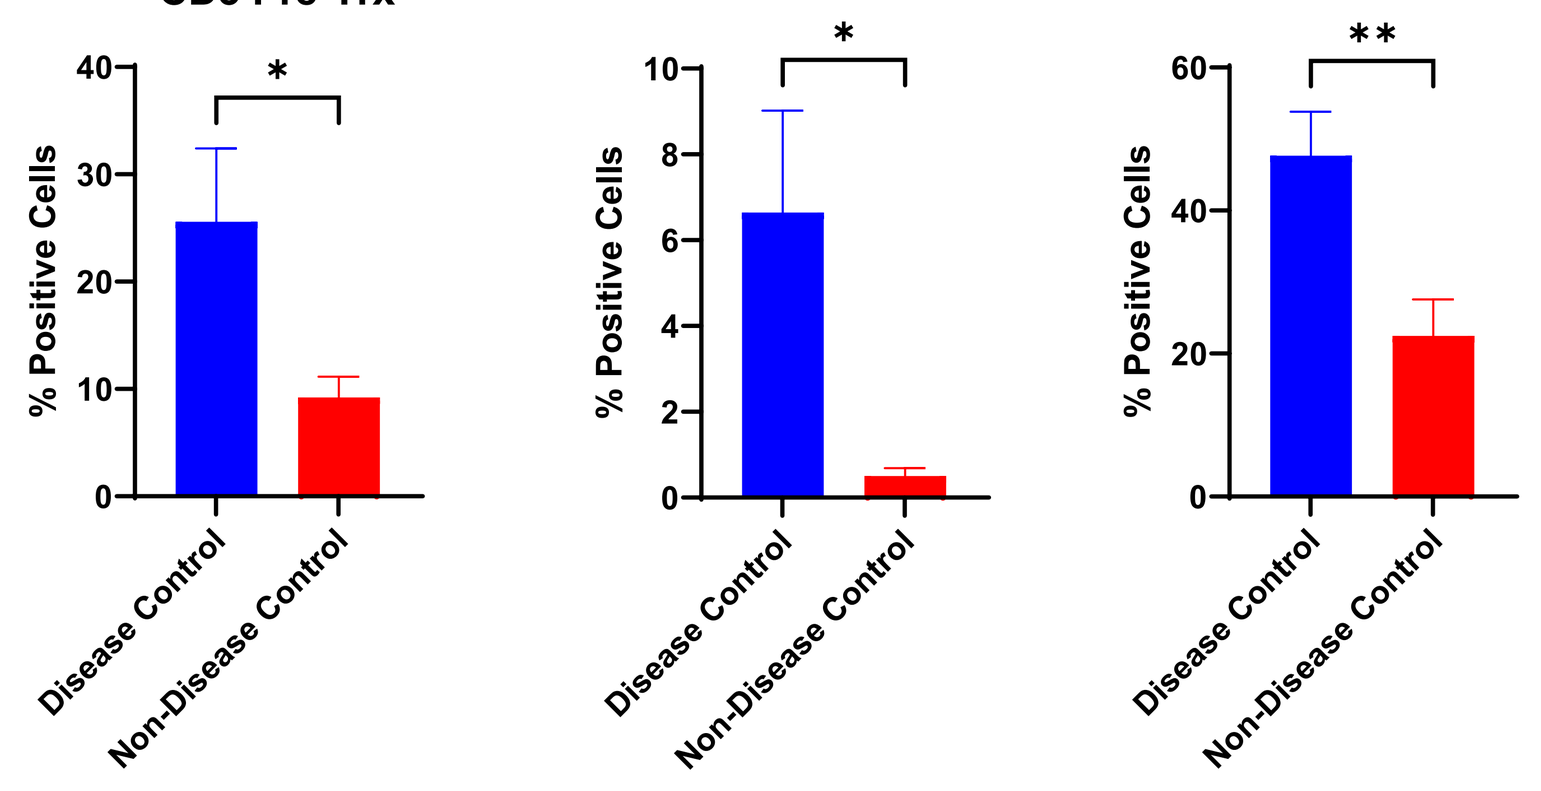

CD3 Pre-Trx
CLEC9A Pre-Trx
CD68 Pre-Trx
% Positive Cells
*
*
**
Disease Control
Non-Disease Control

MEM-288 Increase TME T Cells and Dendritic Cells with Simultaneous Tumor Apoptosis/Necrosis

Pre-Treatment Biopsy

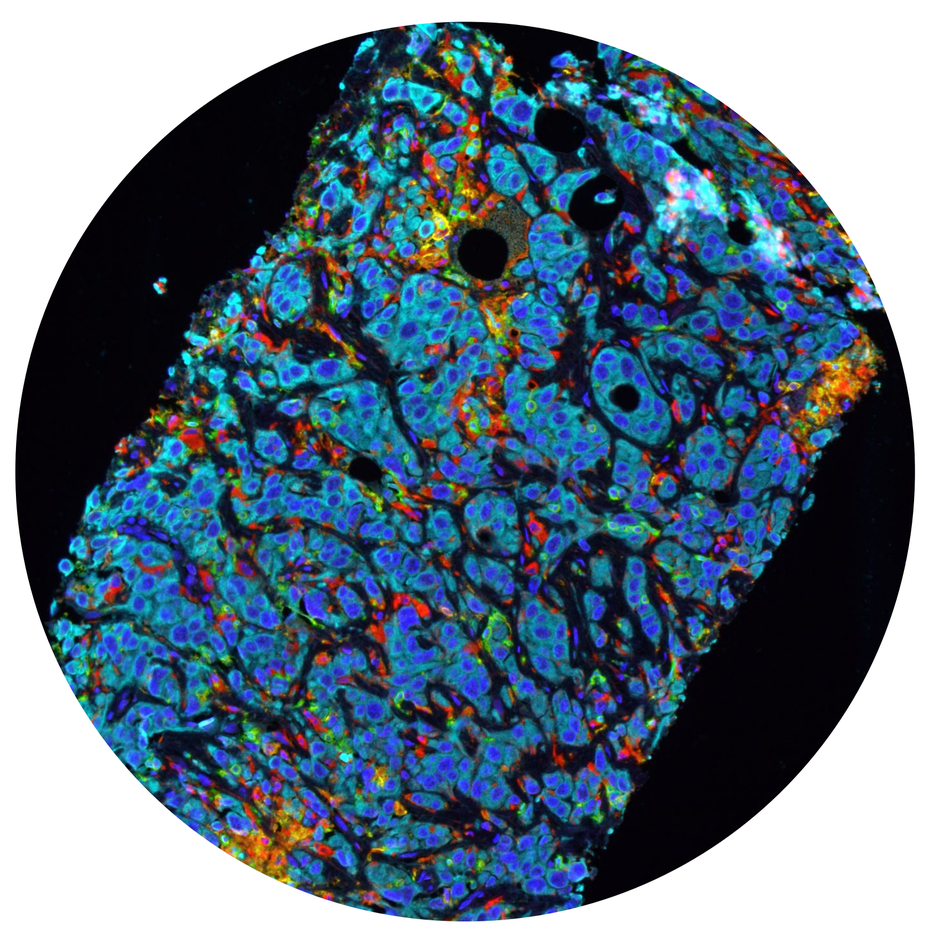

On-Treatment Biopsy

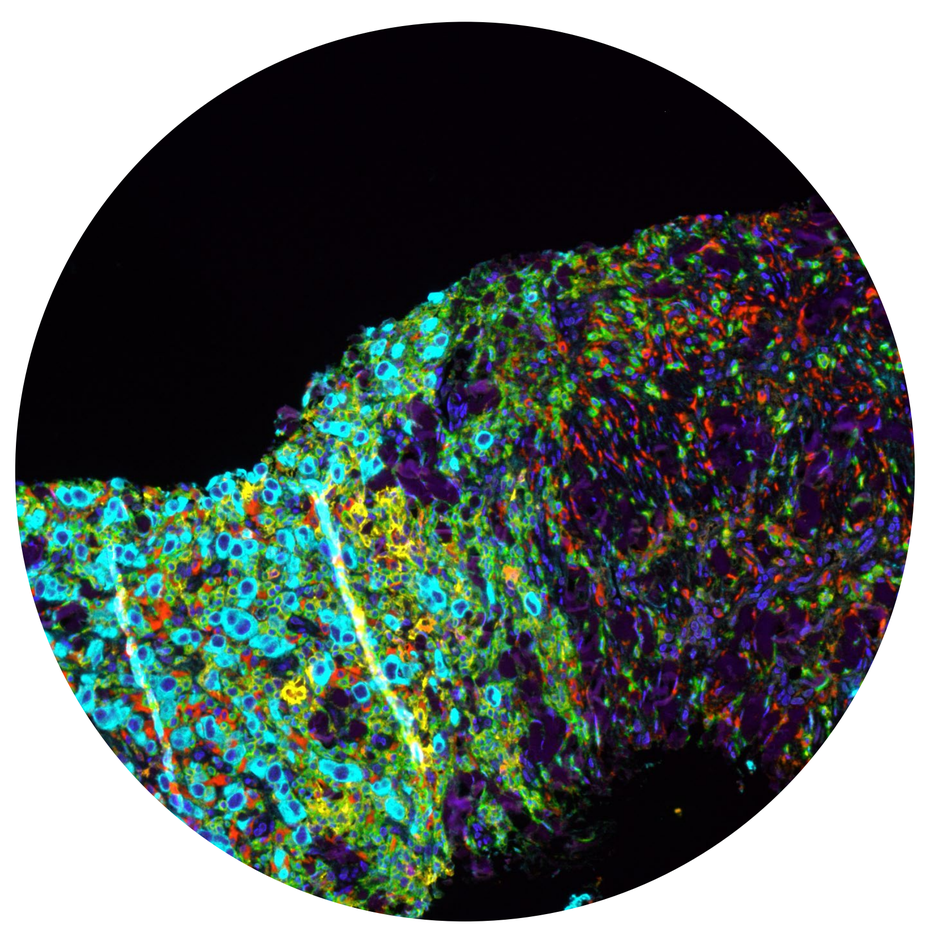

Color Key: Tumor Cells (Blue/Teal) T Cells (Green) Dendritic Cells (Yellow) Macrophages (Red) Stroma (Purple)

MEM-288 Induces Secretion of Multiple Cytokines

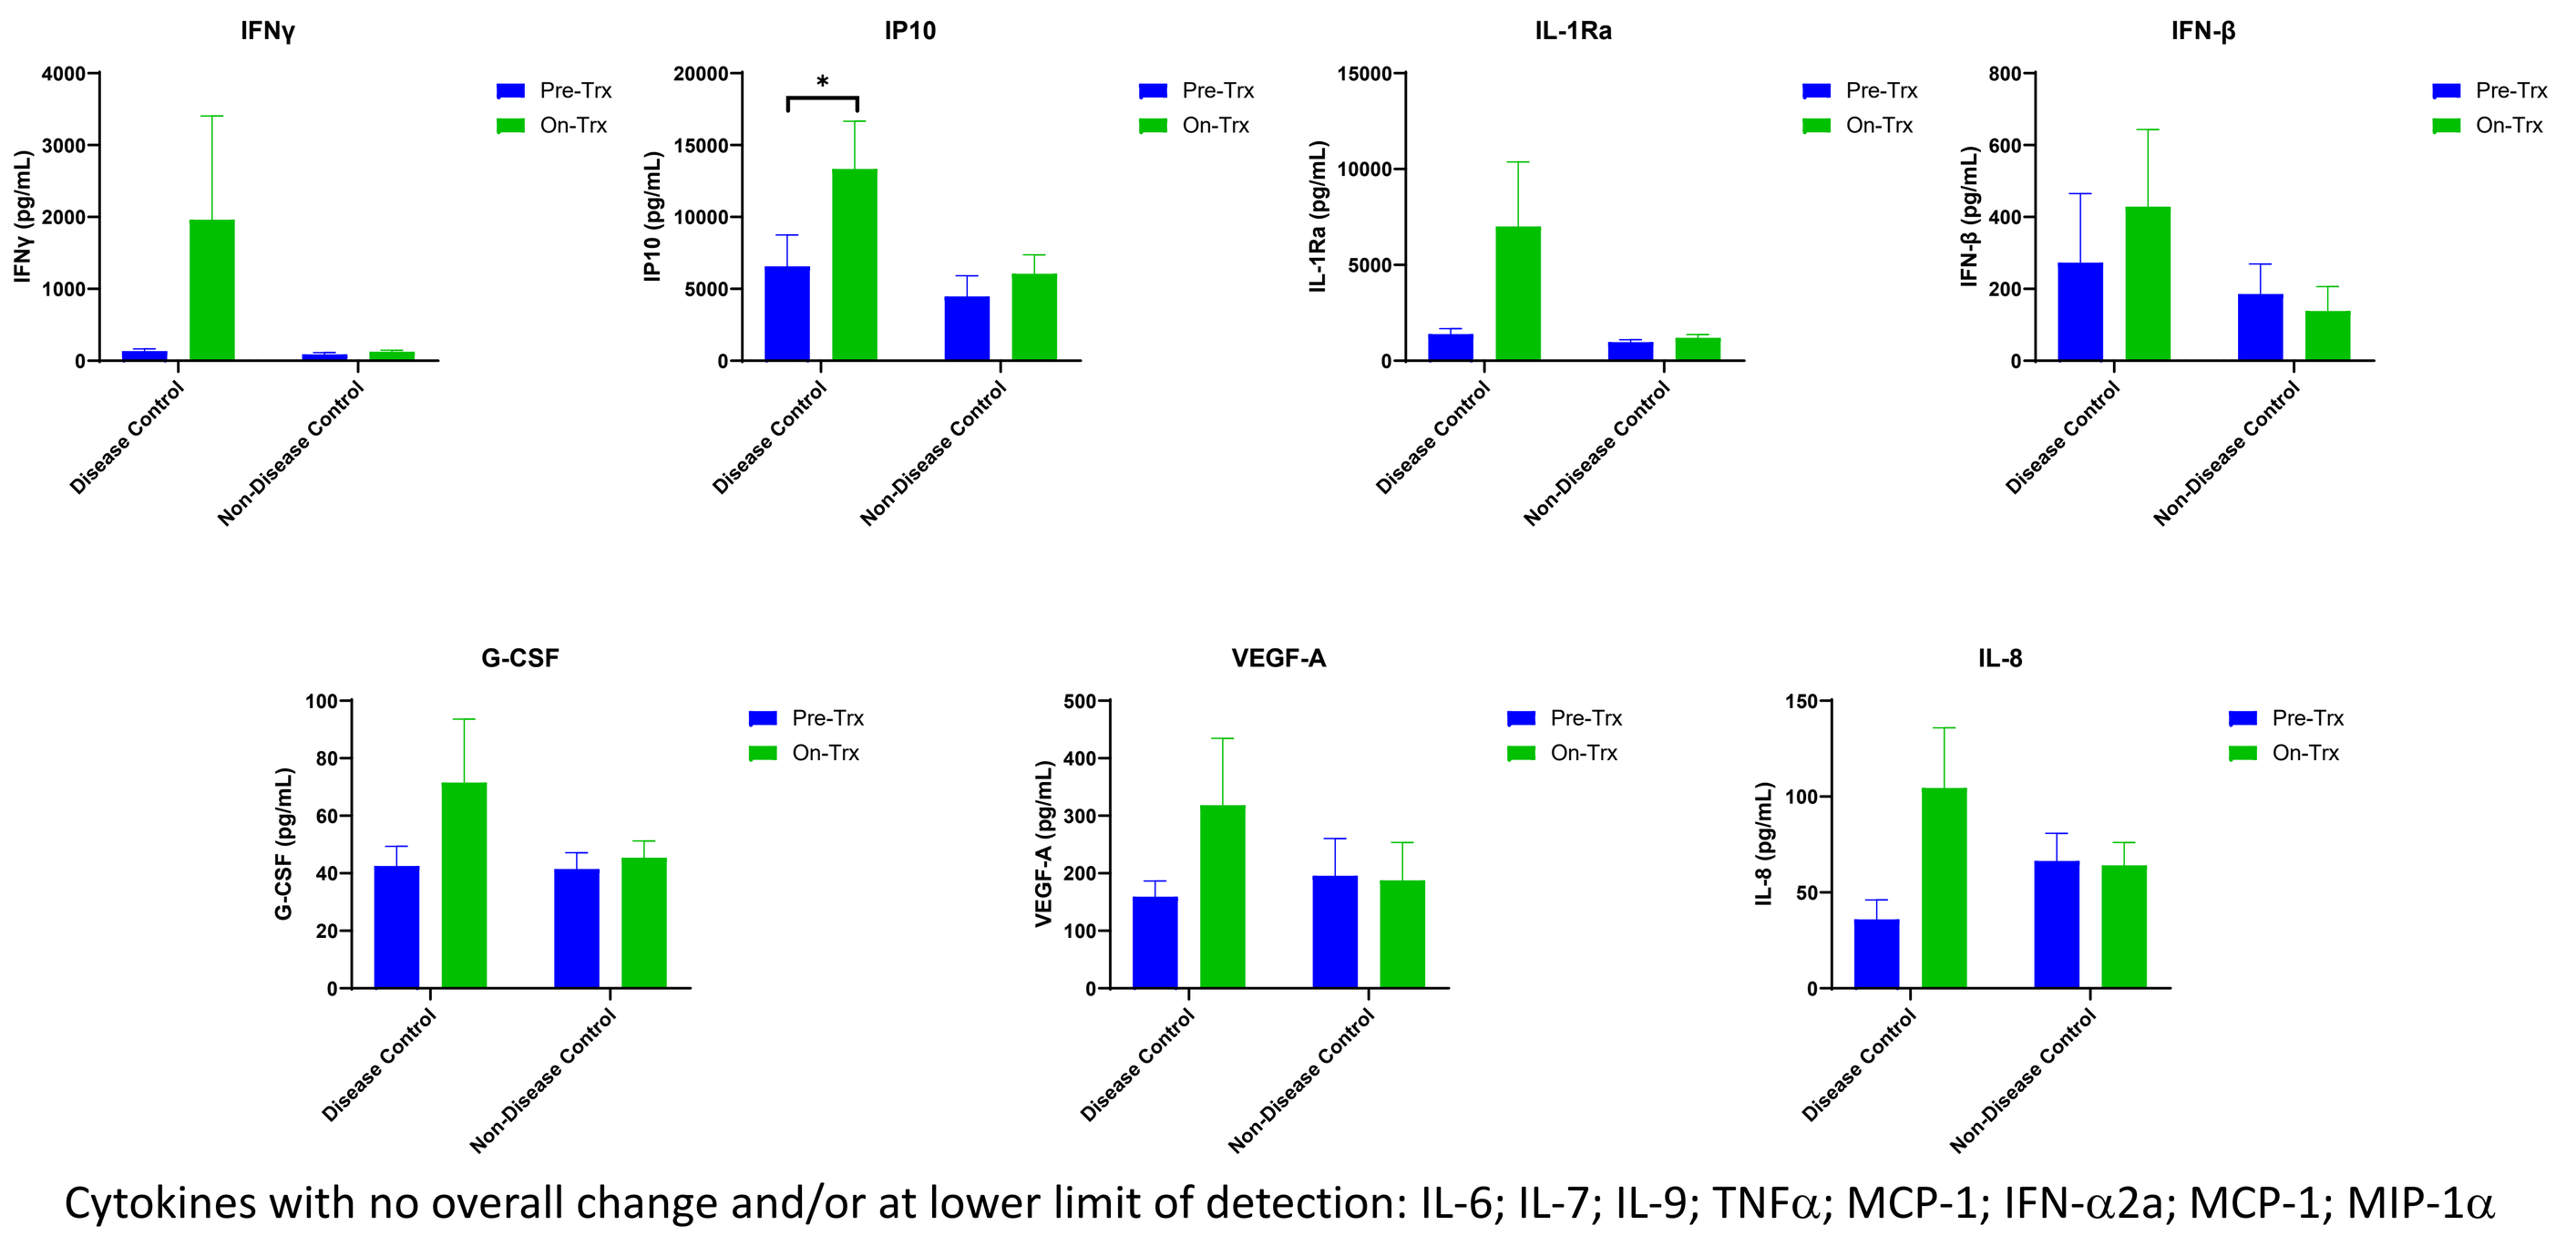

IFNγ
IP10
IL-1Ra
IFN-β
G-CSF
VEGF-A
IL-8
Pre-Trx
On-Trx
Disease Control
Non-Disease Control

Cytokines with no overall change and/or at lower limit of detection: IL-6; IL-7; IL-9; TNFα; MCP-1; IFN-α2a; MCP-1; MIP-1α

Gene Set Enrichment Analysis (GSEA) Shows Enriched Hallmark Pathways after MEM-288 Treatment (on- vs. pre-trx)

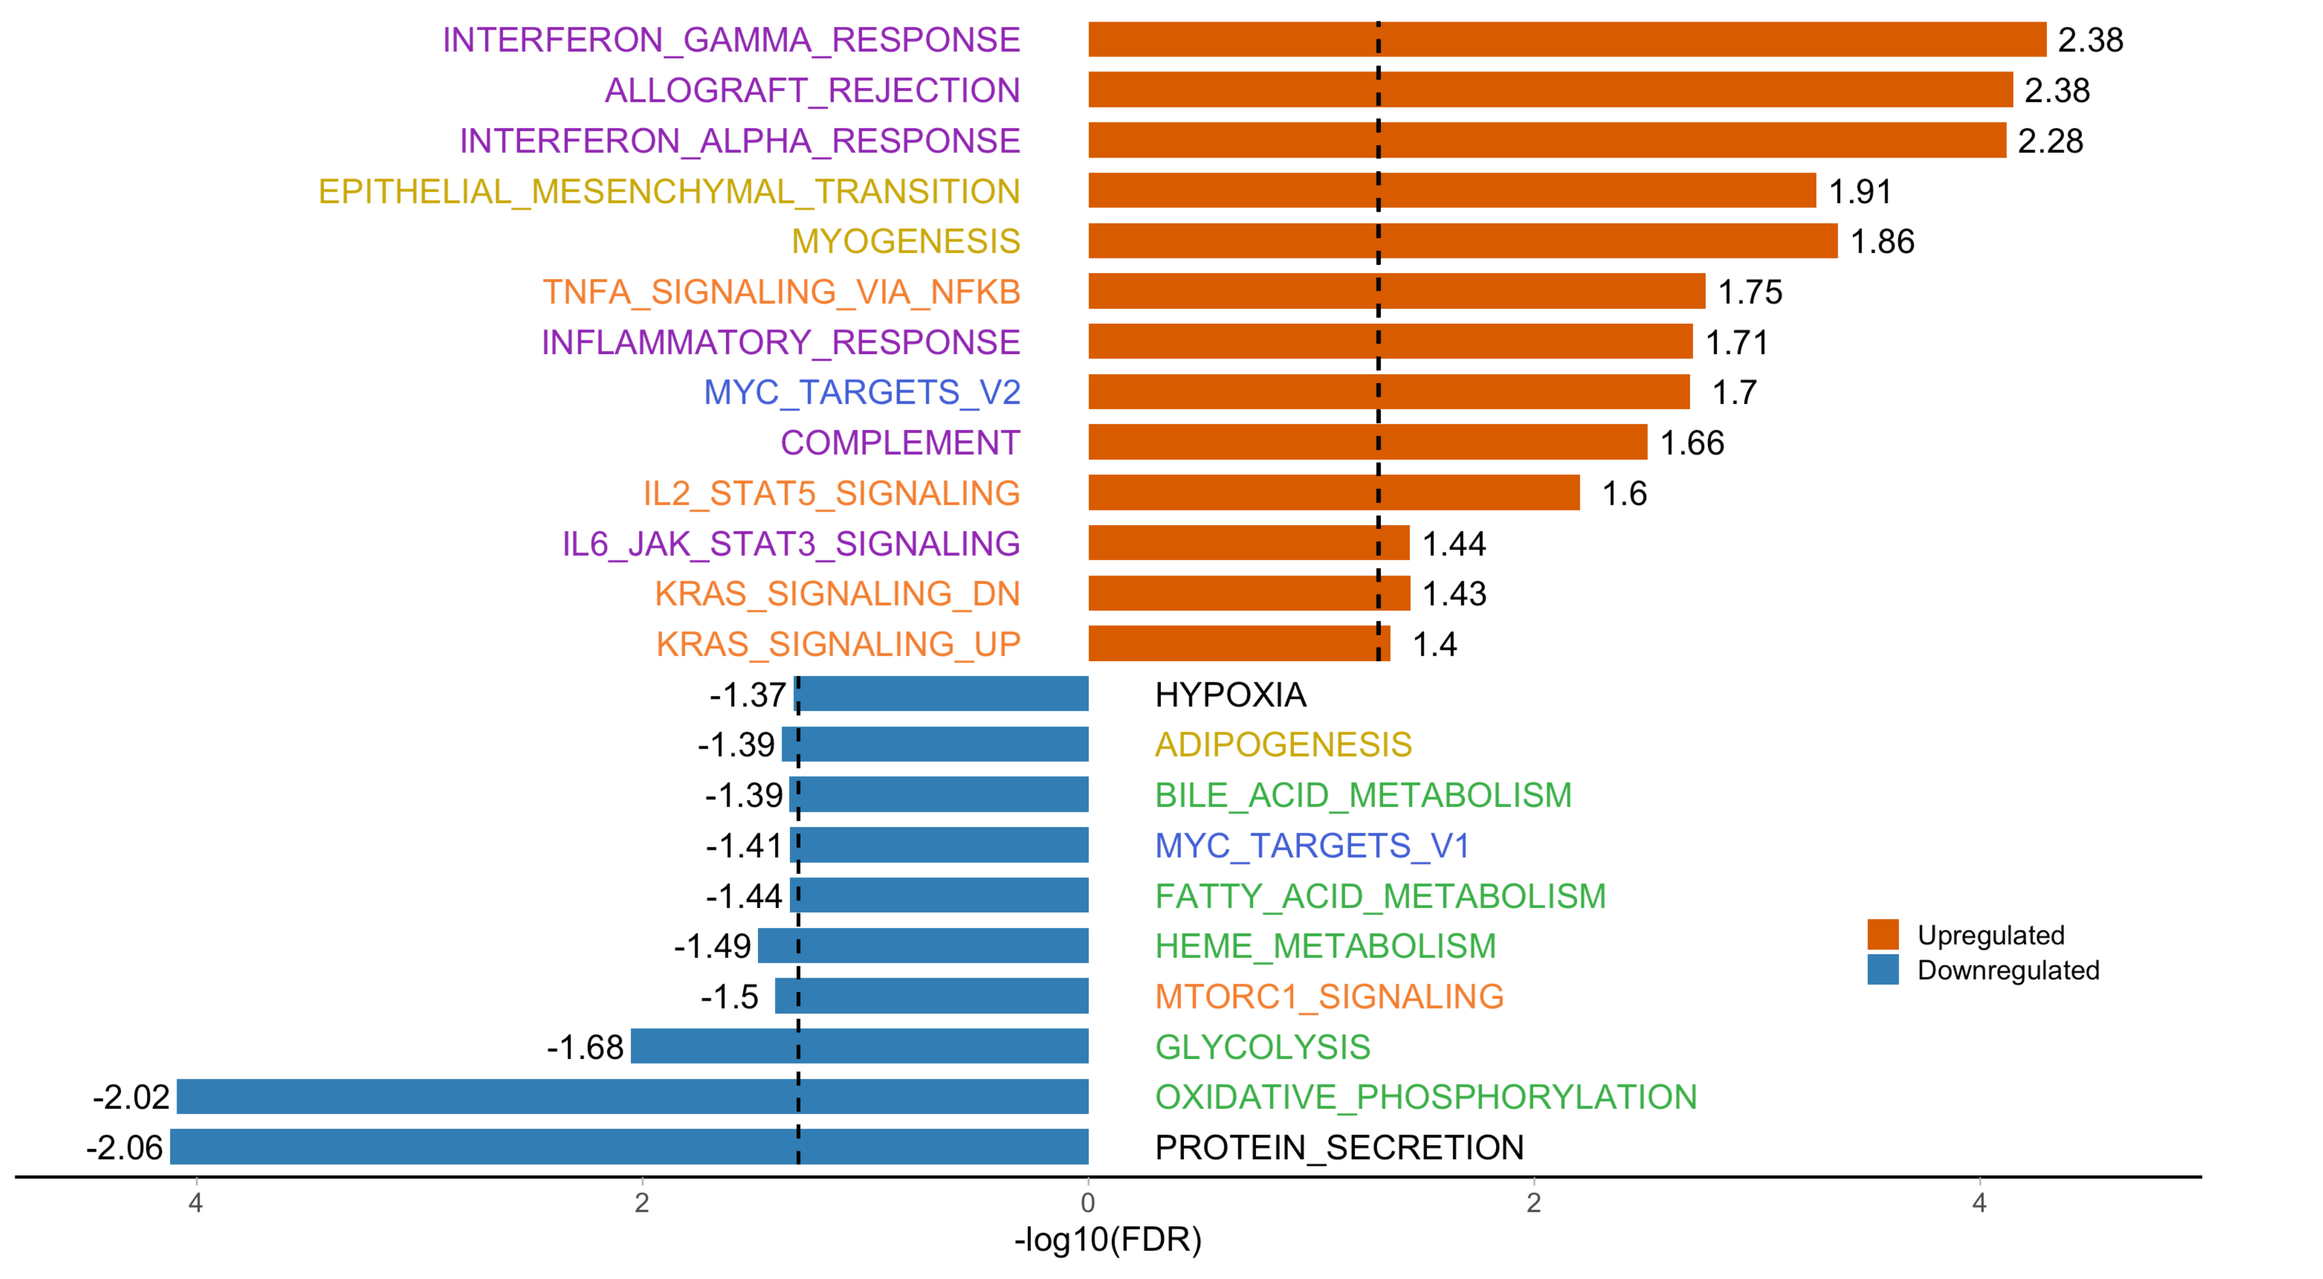

INTERFERON_GAMMA_RESPONSE 2.38
ALLOGRAFT_REJECTION 2.38
INTERFERON_ALPHA_RESPONSE 2.28
EPITHELIAL_MESENCHYMAL_TRANSITION 1.91
MYOGENESIS 1.86
TNFA_SIGNALING_VIA_NFKB 1.75
INFLAMMATORY_RESPONSE 1.71
MYC_TARGETS_V2 1.7
COMPLEMENT 1.66
IL2_STAT5_SIGNALING 1.6
IL6_JAK_STAT3_SIGNALING 1.44
KRAS_SIGNALING_DN 1.43
KRAS_SIGNALING_UP 1.4
HYPOXIA -1.37
ADIPOGENESIS -1.39
BILE_ACID_METABOLISM -1.39
MYC_TARGETS_V1 -1.41
FATTY_ACID_METABOLISM -1.44
HEME_METABOLISM -1.49
MTORC1_SIGNALING -1.5
GLYCOLYSIS -1.68
OXIDATIVE_PHOSPHORYLATION -2.02
PROTEIN_SECRETION -2.06
Upregulated
Downregulated
-log10(FDR)

RNA-Seq analysis demonstrates MEM-288 upregulates multiple gene pathways, including those for interferon responses (correlates with plasma cytokine results) and allograft rejection pathways (surrogate for T cell activation)

MEM-288 Elicits Systemic Immune Responses Against Tumor Neoantigens

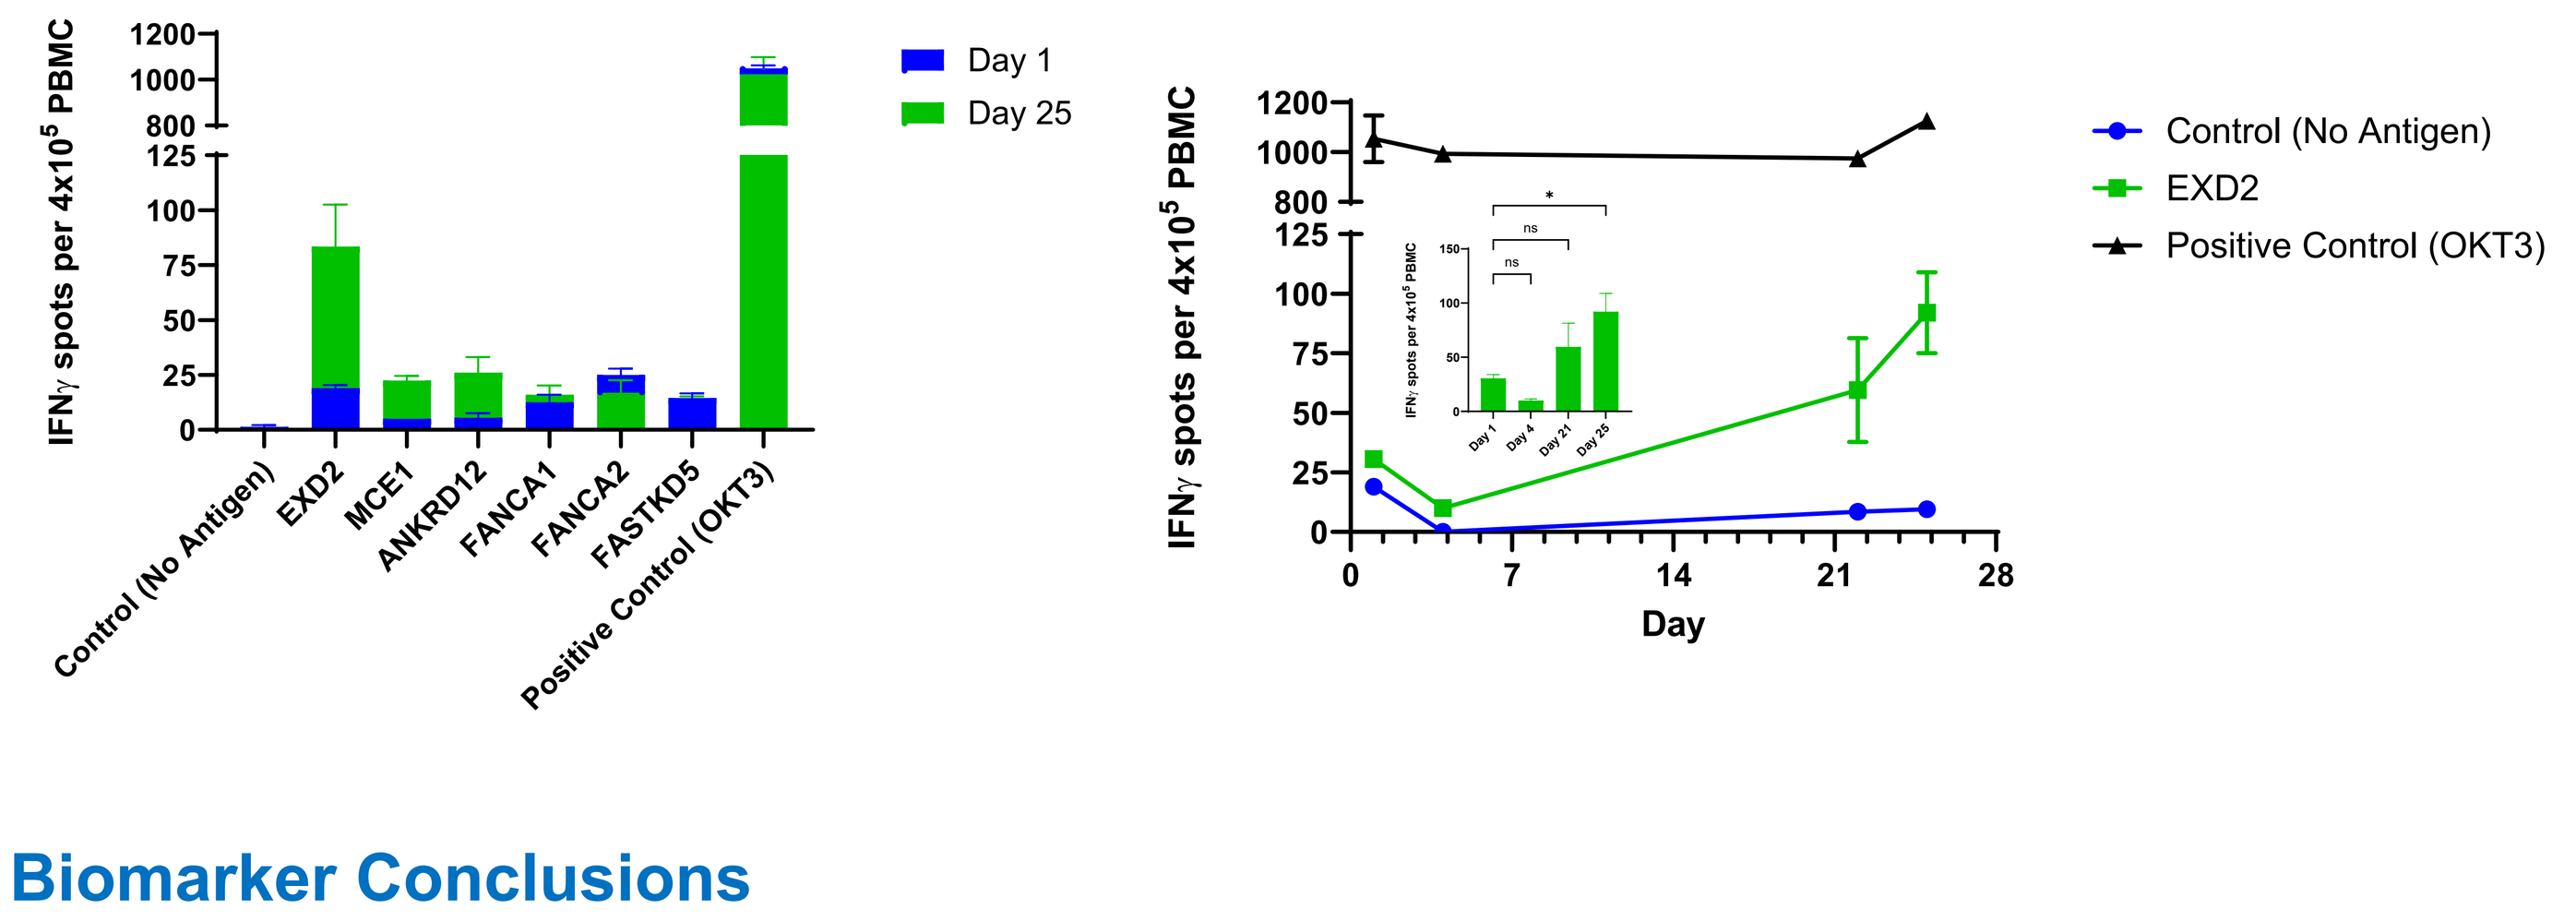

IFNγ spots per 4x10^5 PBMC
Day 1
Day 25
Control (No Antigen)
EXD2
MCE1
ANKRD12
FANCA1
FANCA2
FASTKD5
Positive Control (OKT3)
Control (No Antigen)
EXD2
Positive Control (OKT3)
Day

Biomarker Conclusions

- Pre-treatment presence of T-cells, conventional DC1 cells, and macrophages in tumors significantly (p<0.05) associated with a MEM-288 tumor response
- MEM-288 generated systemic anti-tumor activity in responding patients, included cytokines, TME T cells and dendritic cell increases, gene sets associated with anti-tumor activity, and tumor neoantigen-reactive peripheral T-cells.
- Tumor shrinkage associated with an immune-rich TME in the injected tumors, systemic immune response activation, and strong benefit to chemotherapy rechallenge and long-term disease control

Acknowledgements

- ClinicalTrials.gov Identifier: NCT05076760
- Contact: andreas.saltos@moffitt.org
- Clinical trial funding provided by Memgen, Inc

Overall Conclusions

- Intratumoral injection of MEM-288 is safe and feasibly administered by direct injection to both superficial and visceral lesions. 1x10^{11} viral particles has been selected as the recommended phase 2 dose for clinical expansion studies.
- MEM-288 generated meaningful antitumor activity in advanced NSCLC patients that are refractory to standard therapies including anti-PD(L)1.
- Patient responses were associated with both fewer lines of therapy and pre-treatment TME T cell, dendritic cell, and macrophage presence. MEM-288 induced strong remodeling of the TME, including robust T cell and dendritic cell responses.
- MEM-288 generates systemic anti-tumor immune responses, including cytokine responses, hallmark gene set upregulation, and tumor neoantigen responses.
- Durable patient responses to chemotherapy rechallenge following MEM-288 in two patients was notable and suggests MEM-288 combination with chemotherapy may be promising clinical development approach.
- These results have guided the design of the next stage of this Phase 1 trial, an expansion study of MEM-288 with nivolumab for second-line treatment of advanced NSCLC refractory to anti-PD(L)1 +/- chemotherapy.

SITC 38th Annual Meeting 2023